Effects of High Schizotypy on Control of Eye Movements: Modulation by Antipsychotic Drugs and Nicotine

J.F. William Deakin[3], Anne Schmechtig[1], Colin T. Dourish[2], Kevin J. Craig[2], Gerard R. Dawson[2], Steve C.R. Williams[1], Jane Lees[3], Lawrence Wilkinson[4], Ulrich Ettinger[5,6]

(1) Department of Neuroimaging Sciences, Institute of Psychiatry, King's College London, UK; (2) P1vital, Department of Psychiatry, University of Oxford, Warneford Hospital, Oxford, UK; (3) Neuroscience and Psychiatry Unit, University of Manchester, Manchester, UK;
(4) Department of Psychological Medicine and Neurology, Cardiff University School of Medicine, Cardiff, UK; (5) Department of Psychiatry, Ludwig-Maximilians-University Munich, Germany; (6) Department of Psychology, Ludwig-Maximilians-University Munich, Germany.

Abstract

Background: Oculomotor dysfunction has been well documented within schizophrenia spectrum populations and has been proposed to be a sensitive biomarker for antipsychotic and cognitive enhancing compounds. This study examined the effects of two antipsychotics and nicotine on oculomotor performance and investigated whether individuals with subclinical levels of schizophrenia-like symptoms (called high schizotypy) show a differential profile of drug response to controls.

Methods: In a randomized double blind, placebo-controlled study 233 participants were randomly assigned to one of 4 drug groups (nicotine, risperidone, amisulpride, placebo) and performed prosaccade (PS), antisaccade (AS) and smooth pursuit eye movement (SPEM) tasks. Participants were classified into high and average schizotypy groups measured by the Schizotypal Personality Questionnaire.

Results: Main effects of drug were found for PS gain, PS velocity and AS velocity (all p<0.01), indicating impaired performance with risperidone irrespective of schizotypy level. AS error rate showed a main effect of drug (p<0.01) as well as a drug by group interaction (p=0.035). This pattern indicated that the performance of high schizotypes tended to be worse than controls after placebo but better after antipsychotics. Both groups improved with nicotine treatment. For SPEM a trend for a main effect of drug was found for velocity gain (p=0.052), indicating reduced gain with risperidone irrespective of schizotypy level.

Discussion: In this study eye movement performance proved to be a sensitive measure of pharmacological treatment effects. The main findings were: (1) replication of previous findings of cognitive enhancing effects of nicotine on antisaccade performance; (2) risperidone caused a slowing of AS and PS peak saccade velocity, a reduction in PS spatial accuracy, and an impairment in the ability to match eye velocity to target velocity during SPEM; (3) for the key biomarker of antisaccade error rate, drug effects were modulated as predicted by schizotypy status - risperidone and amisulpride tended to disrupt saccade inhibition in controls but to improve it in high schizotypes. Together, these findings represent promising trends for future research investigating pharmacological influences on high risk and schizophrenia spectrum populations.
This study was supported by the P1vital CNS Experimental Medicine Consortium {members AstraZeneca, GlaxoSmithKline, Lundbeck, Organon (a subsidiary of Merck) and Pfizer}.

Introduction

- The personality trait of schizotypy is a multidimensional continuous construct which has been hypothesized to reflect a subclinical level of schizophrenia and provides insight into the aetiology, treatment and prevention of the disease[1].
- Similarities between schizotypy and schizophrenia have been identified on neuro-cognitive and neuronal levels. An overlap in treatment response characteristics has also been reported[2-4].
- Schizotypy offers a new perspective as a potential model system of drug development for schizophrenia.
- Eye movement deficits have also been observed in schizophrenic patients, their relatives and related spectrum populations[6-7] suggesting that they may be a sensitive biomarker for the disease[6-7].
- Here we combine the model of psychometric schizotypy with the biomarker approach to enhance our understanding of drug response characteristics in the schizophrenia spectrum.

Aims & Objectives

- To examine the effects of antipsychotics and nicotine on oculomotor performance and to investigate whether individuals with subclinical levels of schizophrenia-like symptoms (high schizotypy) show a differential profile of drug response to controls.

Materials & Methods

- 240 healthy volunteers were recruited as part of a multicentre study (Manchester, Cardiff, London)

Recruitment:

- Participants were asked to fill in an online version of Schizotypal Personality Questionnaire (SPQ), suitable participants repeated the SPQ under lab conditions and were divided into medium (between 21-36 -control group: MSz) or high (≥40) scorers (HSz).
- Demographic data was collected and matched between the schizotypy groups (see Table 1).

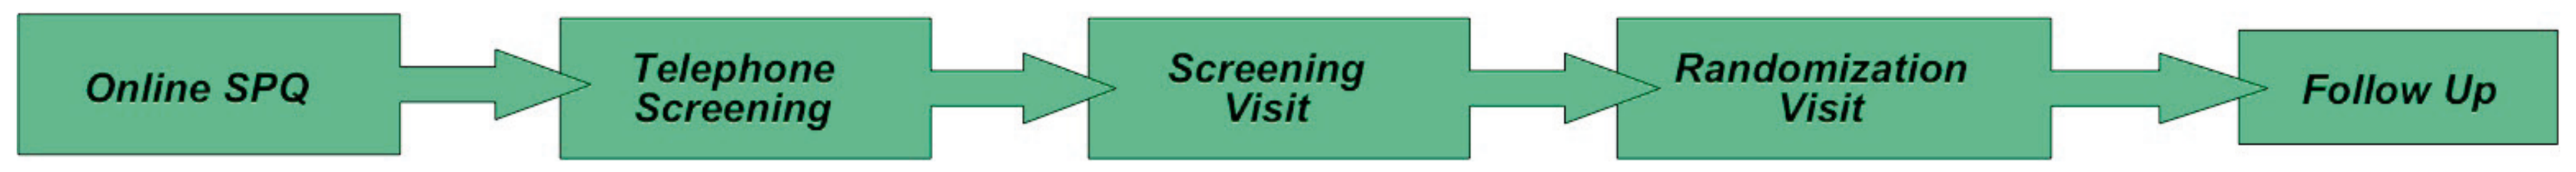
Online SPQ
Telephone Screening
Screening Visit
Randomization Visit
Follow Up

- Standard screening methods and exclusion criteria for pharmacological studies were applied on screening and randomization day.

Study design:

- Double-blind, randomised, placebo controlled, parallel study design with 4 study arms:
 - Placebo (PLA)
 - Risperidone- 2mg (RIS)
 - Amisulpride- 400mg (AMI)
 - Nicotine- 7mg (NIC)
- Focal imaging technology (Eyelink 1000 - SR Research Ltd., Toronto-Ottawa, Canada) with sampling rate of 1000Hz frequency was used to record eye tracking

Tasks:

- Four possible peripheral positions of a black dot on white background (±7.25°,±14.5°) for the random duration of 1000-2000ms, 60 trials in total

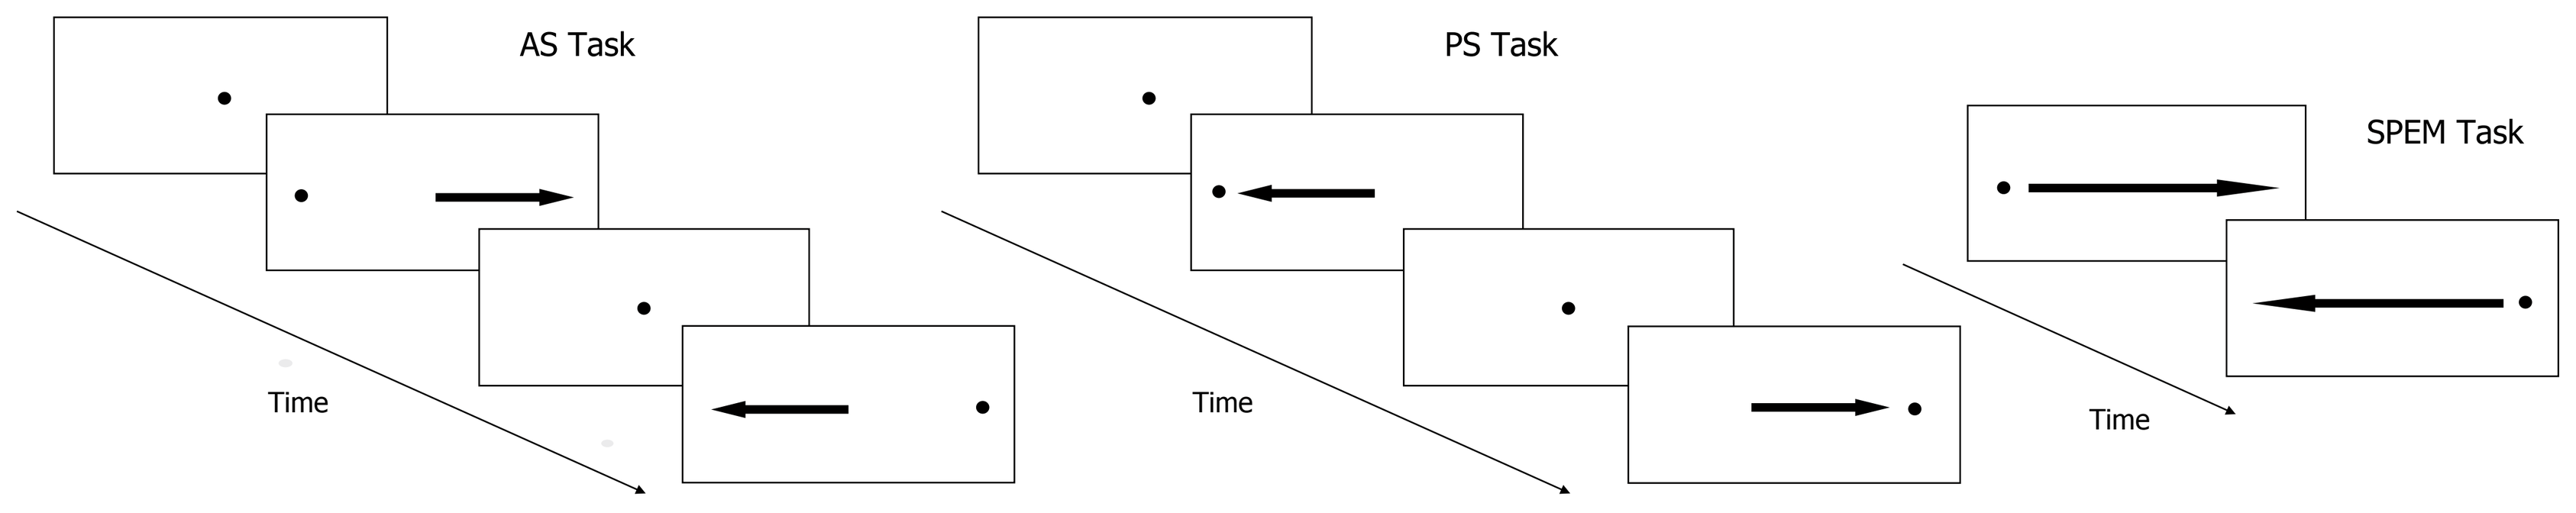
AS Task
Time
PS Task
Time
SPEM Task
Time

Dependent variables:

- Antisaccade (AS)/ Prosaccades (PS): directional error rate, latency, amplitude gain, peak velocity
- Smooth Pursuits task (SPEM): sinusoidal target velocities: 12°/s, 24°/s, 36°/s, 20 half-cycles: SPEM velocity gain, SPEM saccadic frequency

Analyses

- AS/PS: Eye Link Data Viewer (SR Research Ltd., Mississauga, Ontario, Canada)
- SPEM: LabView (National Instruments Corporation)
- Statistical analysis was carried out using univariate/repeated measure ANOVA models in SPSS 15.0 (SPSS Inc., Chicago, IL, USA)
 - Within-subject factor: Saccade Task (AS, PS) or SPEM Task (3 velocities: 12°/s, 24°/s, 36°/s)
 - Between-subject factors: Site, Drug, Schizotypy Group, Gender
 - Covariates: Age, IQ, Time since dosing (included if significant)
- Post-hoc comparisons were performed using Bonferroni corrections

Results

Table 1: Descriptive statistics for demographic variables

	Schizotypy level		Statistical Test
	Medium	High	
Age in Years	23.56 (5.71)	23.9 (5.31)	F [1,229]=0.21, p=0.64
[range]	[18-42]	[18-44]	
IQ (NART score)	113.38 (4.99)	113.58 (5.17)	F[1,231]=0.08, p=0.77
N male (%)	59 (25)	57 (24)	X^2=0.04, df=1, p=0.90
N right-handed	104	105	X^2=2.8, df=1, p=0.25
Ethnicity (%)			
Caucasian	104 (45)	89 (38)	X^2=6.24, df=1, p=0.10
Asian	10 (4)	19 (8)	
Black	2 (1)	5 (2)	
Other	1 (0.5)	3 (1)	

Note: Data represent means (SDs) by schizotypy group unless otherwise stated

Figure 1:
This shows Antisaccade Error Rate by Schizotypy Group. Bars represent Means, whiskers represent 95% Confidence Interval for Mean.

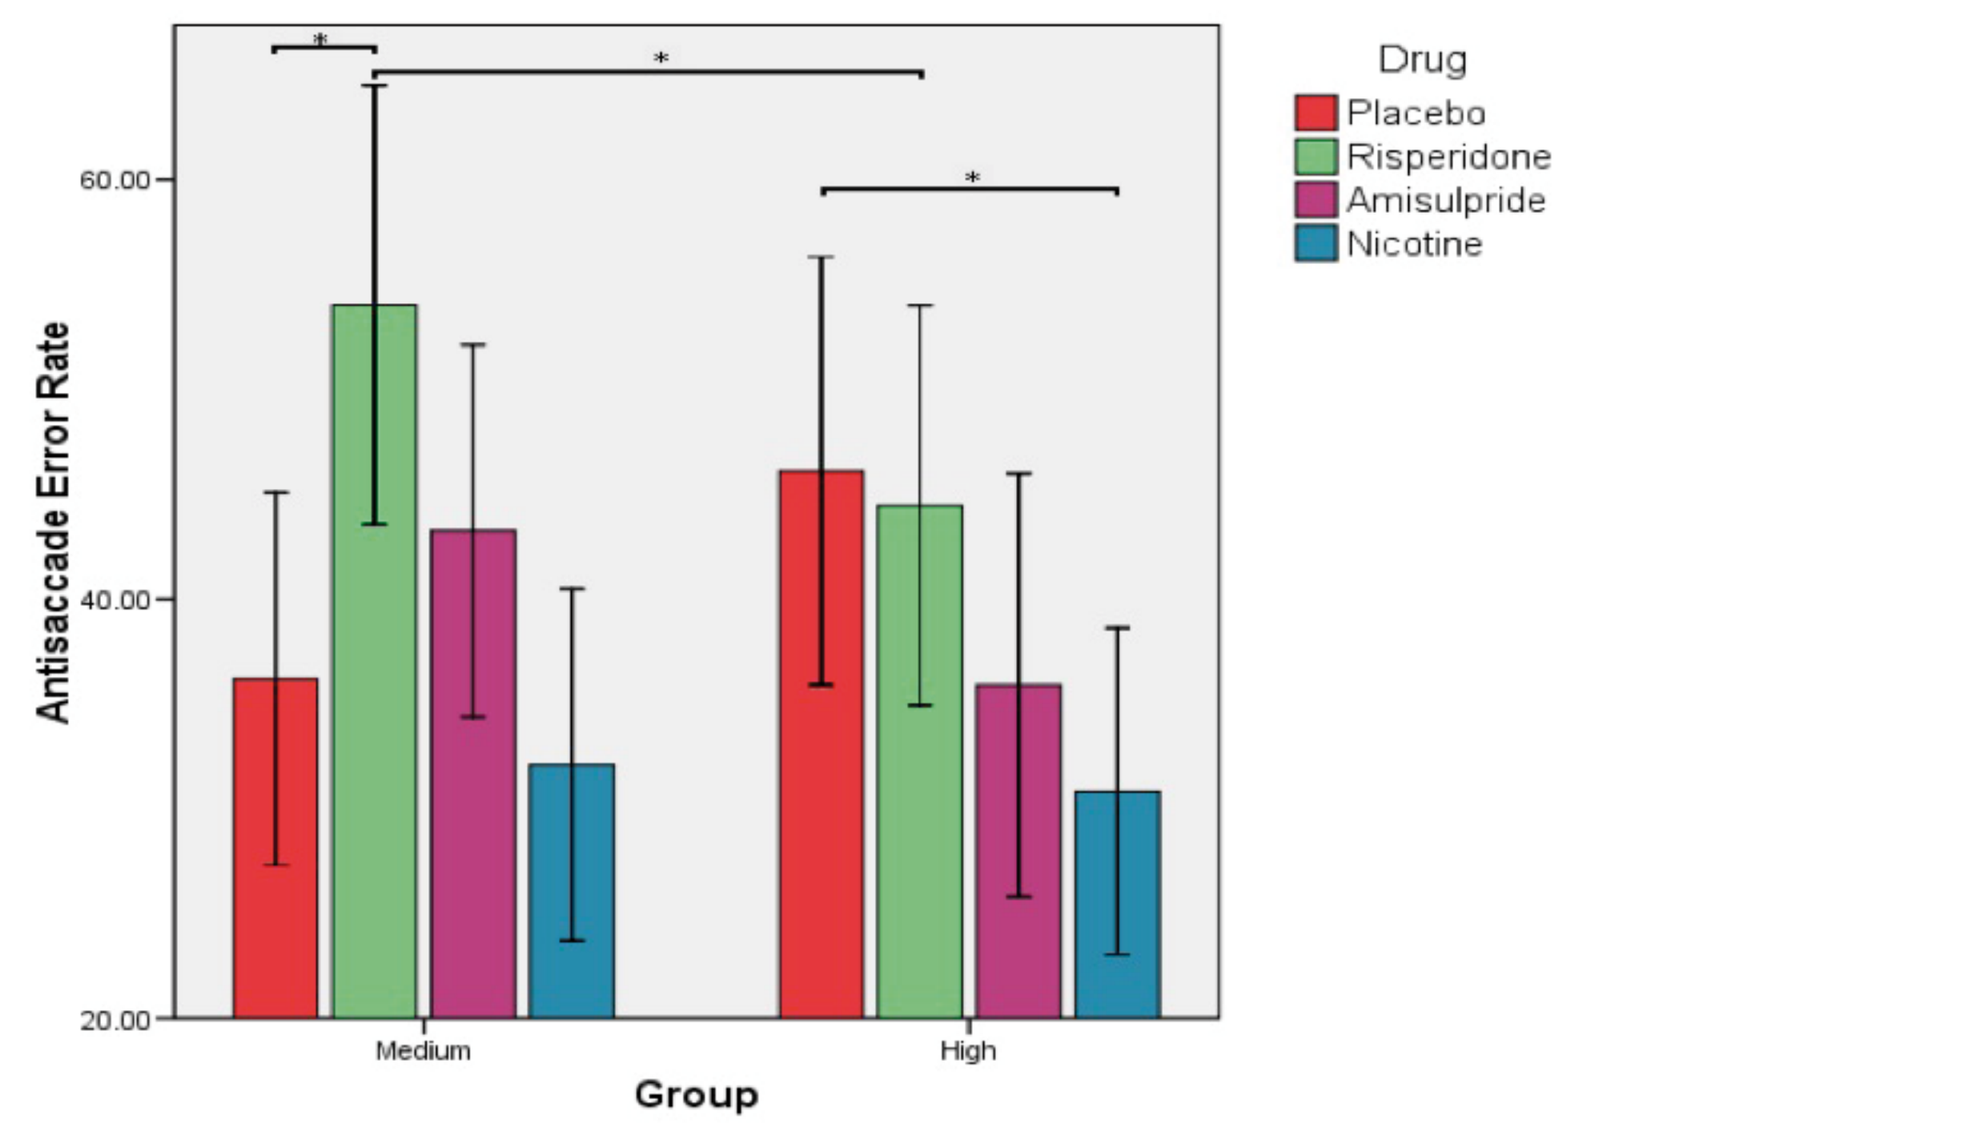
Drug
Placebo
Risperidone
Amisulpride
Nicotine
Antisaccade Error Rate
60.00
40.00
20.00
Medium
High
Group

Figure 2:
This shows Estimated Marginal Means of Saccadic Peak Velocity by Drug Group irrespective of Schizotypy Level.

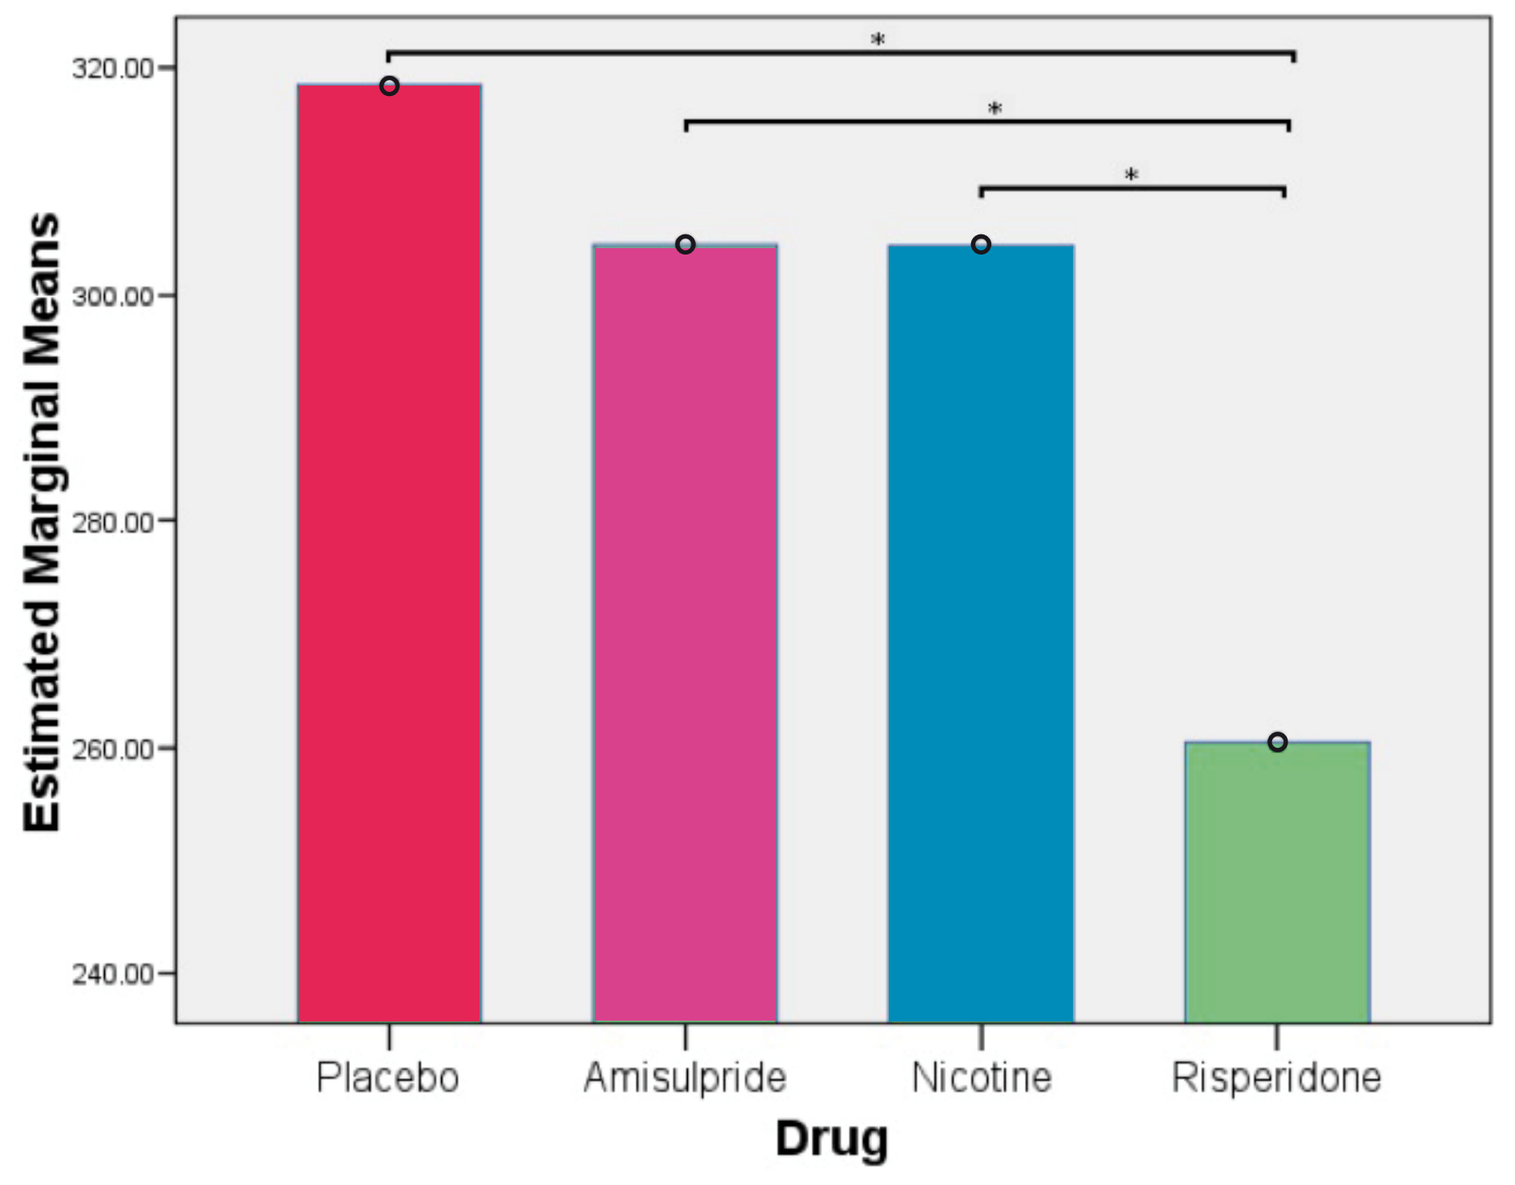
Estimated Marginal Means
320.00
300.00
280.00
260.00
240.00
Placebo
Amisulpride
Nicotine
Risperidone
Drug

- For the antisaccade error rate a significant main effect of drug (F(3,179)=5.86, p<0.01) as well as a drug by group interaction (F(3,179)=2.97, p=0.03) was found indicating a trend that HSz performance was worse than MSz on PLA but improved with both antipsychotics. Both groups profited from NIC (p=0.04). HSz significantly improved in performance on NIC compared to PLA (p=0.02). In the MSz group RIS significantly worsened performance compared to PLA (p=0.03). The MSz group performed significantly worse on RIS compared to the HSz group (p=0.04). See Figure 1 for details.
- A main effect of task (F(1,183)=97.69, p<0.01) and drug (F(3,183)=12.06, p<0.01) was found for peak velocity with RIS impairing performance compared to PLA, AMS and NIC irrespective of schizotypy group (all p<0.01). See Figure 2 for details.
- SPEM gain showed a main effect of task (F(2,350)=13.36, p<0.01) as well as a trend for a drug effect (F(3,175)=2.38, p=0.07) with RIS displaying the tendency to impair the ability to match eye velocity to target velocity compared to NIC (p=0.05).
- No main effect of schizotypy group was observed in this study. A main effect of site was found for AS error rate (F(2,179)=7.52, p<0.01) indicating lower error rates in Cardiff compared to Manchester and London. Also a significant effect of gender (F(1,175)=4.31, p=0.04) was found for SPEM gain with males performing significantly better than females.

Discussion

- We were able to replicate previous findings of the cognition enhancing effects of NIC on antisaccade performance[8].
- RIS caused a slowing of peak saccade velocity and an impairment in the ability to match eye velocity to target velocity during SPEM was found reflecting its sedative properties[9]
- For the key biomarker of antisaccade error rate there was a trend towards the expected modulation of drug effects by schizotypy status: RIS and AMI tended to disrupt saccade inhibition in MSz but improve it in HSz. The HSz improved more on NIC indicating a higher sensitivity to the influence of cholinergic pharmaceuticals potentially remediating associated cognitive deficits in an imbalanced cholinergic system.
- The schizotypy model has been found to be suitable for drug investigations. There are no overt symptoms and illness- or treatment-related confounds in this schizophrenia spectrum population allowing for confounder free interpretation of the results.
- Eye movement measures show a high sensitivity to pharmacological interventions. They can be measured objectively and they are easy to apply.
- The findings of this study provide promising leads for future research investigating pharmacological influences on high risk and schizophrenia spectrum populations.

Conclusion

- Multicentre cognitive biomarker studies are feasible and can be reliable
- Antisaccade task sensitive to nicotine especially in high schizoptypes
- Sample selection by schizotypy may amplify the detection of new cognition enhancing agents for schizophrenia

Acknowledgements

This study was supported by the P1vital CNS Experimental Medicine Consortium {members: AstraZeneca, GlaxoSmithKline, Lundbeck, Organon (a subsidiary of Merck) and Pfizer}. Reprint requests to: Anne.Schmechtig@kcl.ac.uk

Disclosures:

A. Schmechtig: None. J. Lees: None. G. Dawson: Part 1; P1vital. Part 2; P1vital. Part 3; P1vital. Part 5; P1vital. C. Dourish: Part 1; P1vital. Part 2; P1vital. Part 3; P1vital. Part 5; P1vital. K. Craig: Part 1; P1vital. Part 2; P1vital. Part 3; P1vital. Part 5; P1vital. B. Deakin: Part 1; P1vital, Servier, Janssen, AstraZeneca. Part 4; P1vital, Servier, AstraZeneca. L. Wilkinson: Part 1; P1vital. Part 4; P1vital, Lilly (UK). S. Willliams: ; P1vital. U. Ettinger: ; P1vital, GE Healthcare.

References

1. van Os J, Linscott RJ, Myin-Germeys I, Delespaul P, Krabbendam L (2009) A systematic review and meta-analysis of the psychosis continuum: evidence for a psychosis proneness-persistence-impairment model of psychotic disorder. Psychol.Med. 39: 179-195;
2. Vollema MG, Postma B (2002) Neurocognitive correlates of schizotypy in first degree relatives of schizophrenia patients. Schizophr.Bull. 28: 367-377;
3. Dickey CC, Shenton ME, Hirayasu Y, Fischer I, Voglmaier MM, Niznikiewicz MA, Seidman LJ, Fraone S, McCarley RW (2000) Large CSF volume not attributable to ventricular volume in schizotypal personality disorder. Am.J.Psychiatry. 157: 48-54;
4. O'Flynn K, Gruzelier JH, Bergman AJ, Siever LJ (2003) The schizophrenia spectrum personality disorders. In: Hirsch SR, Weinberger DR (eds) Schizophrenia. Blackwell, Chichester, pp 80-100;
5. de Visser SJ, van der PJ, Pieters MS, Cohen AF, van Gerven JM (2001) Biomarkers for the effects of antipsychotic drugs in healthy volunteers. Br.J.Clin.Pharmacol. 51: 119-132;
6. Hutton SB, Ettinger U (2006) The antisaccade task as a research tool in psychopathology: a critical review. Psychophysiology 43: 302-313;
7. O'Driscoll GA, Callahan BL (2008) Smooth pursuit in schizophrenia: a meta-analytic review of research since 1993. Brain Cogn. 68: 359-370;
8. Reilly JL, Lencer R, Bishop JR, Keedy S, Sweeney JA (2008) Pharmacological treatment effects on eye movement control. Brain Cogn. 68: 415-435;
9. Barrett SL, Bell R, Watson D, King DJ (2004) Effects of amisulpride, risperidone and chlorpromazine on auditory and visual latent inhibition, prepulse inhibition, executive function and eye movements in healthy volunteers. J.Psychopharmacol. 18: 156-172;